5873

BAY-3827, a selective inhibitor of AMPK for the evaluation of the role of AMPK in MYC-dependent tumors

Clara Lemos[1], Volker K. Schulze[1], Benjamin Bader[1], Clara D. Christ[1], Hans Briem[1], Oliver Politz[1], Florian Prinz[1], Simon Holton[1], Ulf Bömer[1], Tobias Heinrich[1], Julien Lefranc[1], Philip Lienau[1], Arne Scholz[1], Franz von Nussbaum[2], Carl Friedrich Nising[1], Dominik Mumberg[1], Marcus Bauser[1], Andrea Hägebarth[1]

[1]Bayer AG, Pharmaceutical Division, Berlin, Germany; [2]Bayer S.A.S., Crop Science Division, Lyon, France

INTRODUCTION

- AMP-activated protein kinase (AMPK) is a sensor of the energy status in cells and it plays a key role in controlling cell metabolism.
- AMPK is a component of the liver kinase B1 (LKB1) tumor suppressor cascade, which inhibits mammalian target of rapamycin complex 1 (mTORC1) and has mainly been perceived as a tumor suppressor. However, in the last few years, studies have suggested that AMPK might actually exert a pro-tumorigenic role in certain contexts.[1,2]
- MYC is a proto-oncogene upregulated in many forms of cancer. Dysregulated MYC expression has been demonstrated to render tumor cells sensitive to AMPK depletion. Due to their increased anabolism, MYC-dependent cells rely on AMPK to restore ATP levels and to prevent an energy crisis resulting in apoptosis and cell death (**Fig. 1**).[3]
- After target validation using siRNA, our aim was to develop potent and selective AMPK inhibitors to evaluate the therapeutic potential of inhibiting AMPK in MYC-driven tumors.

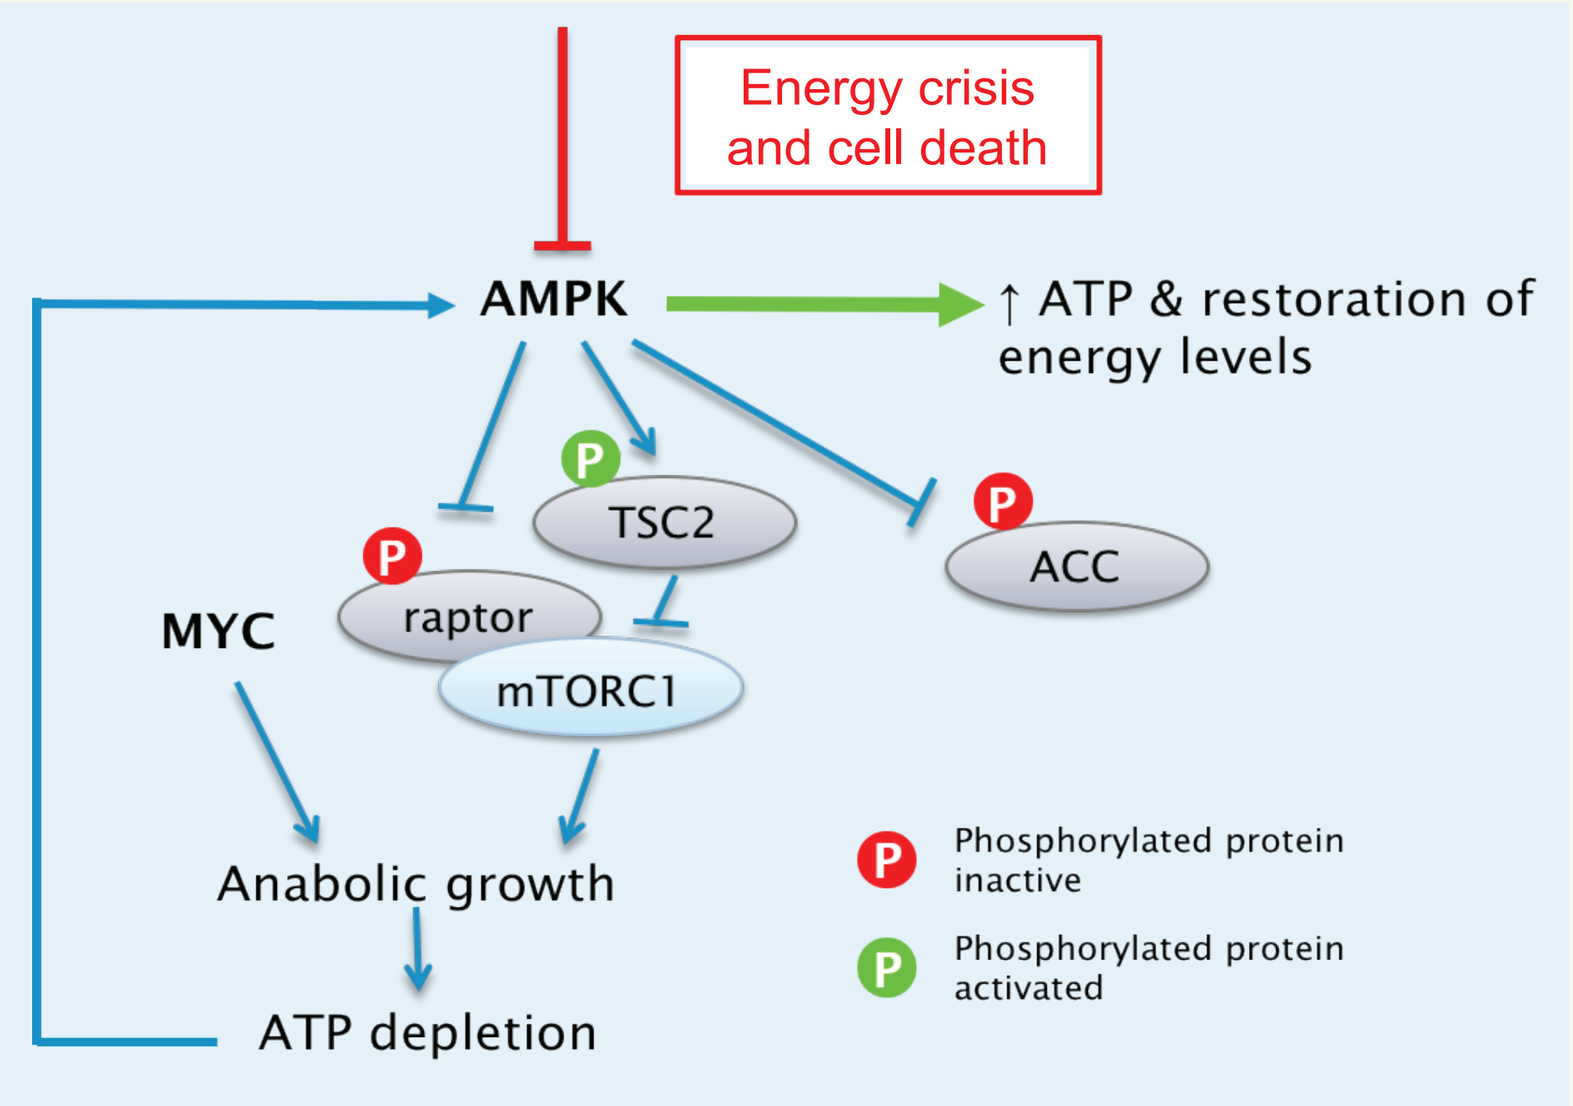
Energy crisis and cell death
AMPK
↑ ATP & restoration of energy levels
TSC2
ACC
raptor
mTORC1
MYC
Anabolic growth
ATP depletion
P Phosphorylated protein inactive
P Phosphorylated protein activated

Figure 1. Target rationale for inhibiting AMPK in MYC-dependent cells. MYC dysregulation triggers increased anabolic activity, resulting in ATP depletion. As a result, AMPK is activated to reduce anabolic activities and to restore cellular energy levels via inactivation of mTORC1. In MYC-dependent cells, the inhibition of AMPK results in the activation of mTORC1 and subsequent anabolic effects, leading to an energy crisis and induction of apoptosis (scheme based on Liu *et al*, 2012).[3] ACC, acetyl-CoA carboxylase; AMPK, 5' adenosine monophosphate-activated protein kinase; ATP, adenosine triphosphate; mTORC1, mammalian target of rapamycin complex 1; TSC2, tuberous sclerosis complex 2.

RESULTS

Structure-activity relationship (SAR) analysis of the high-throughput screening (HTS) hits and first analogues

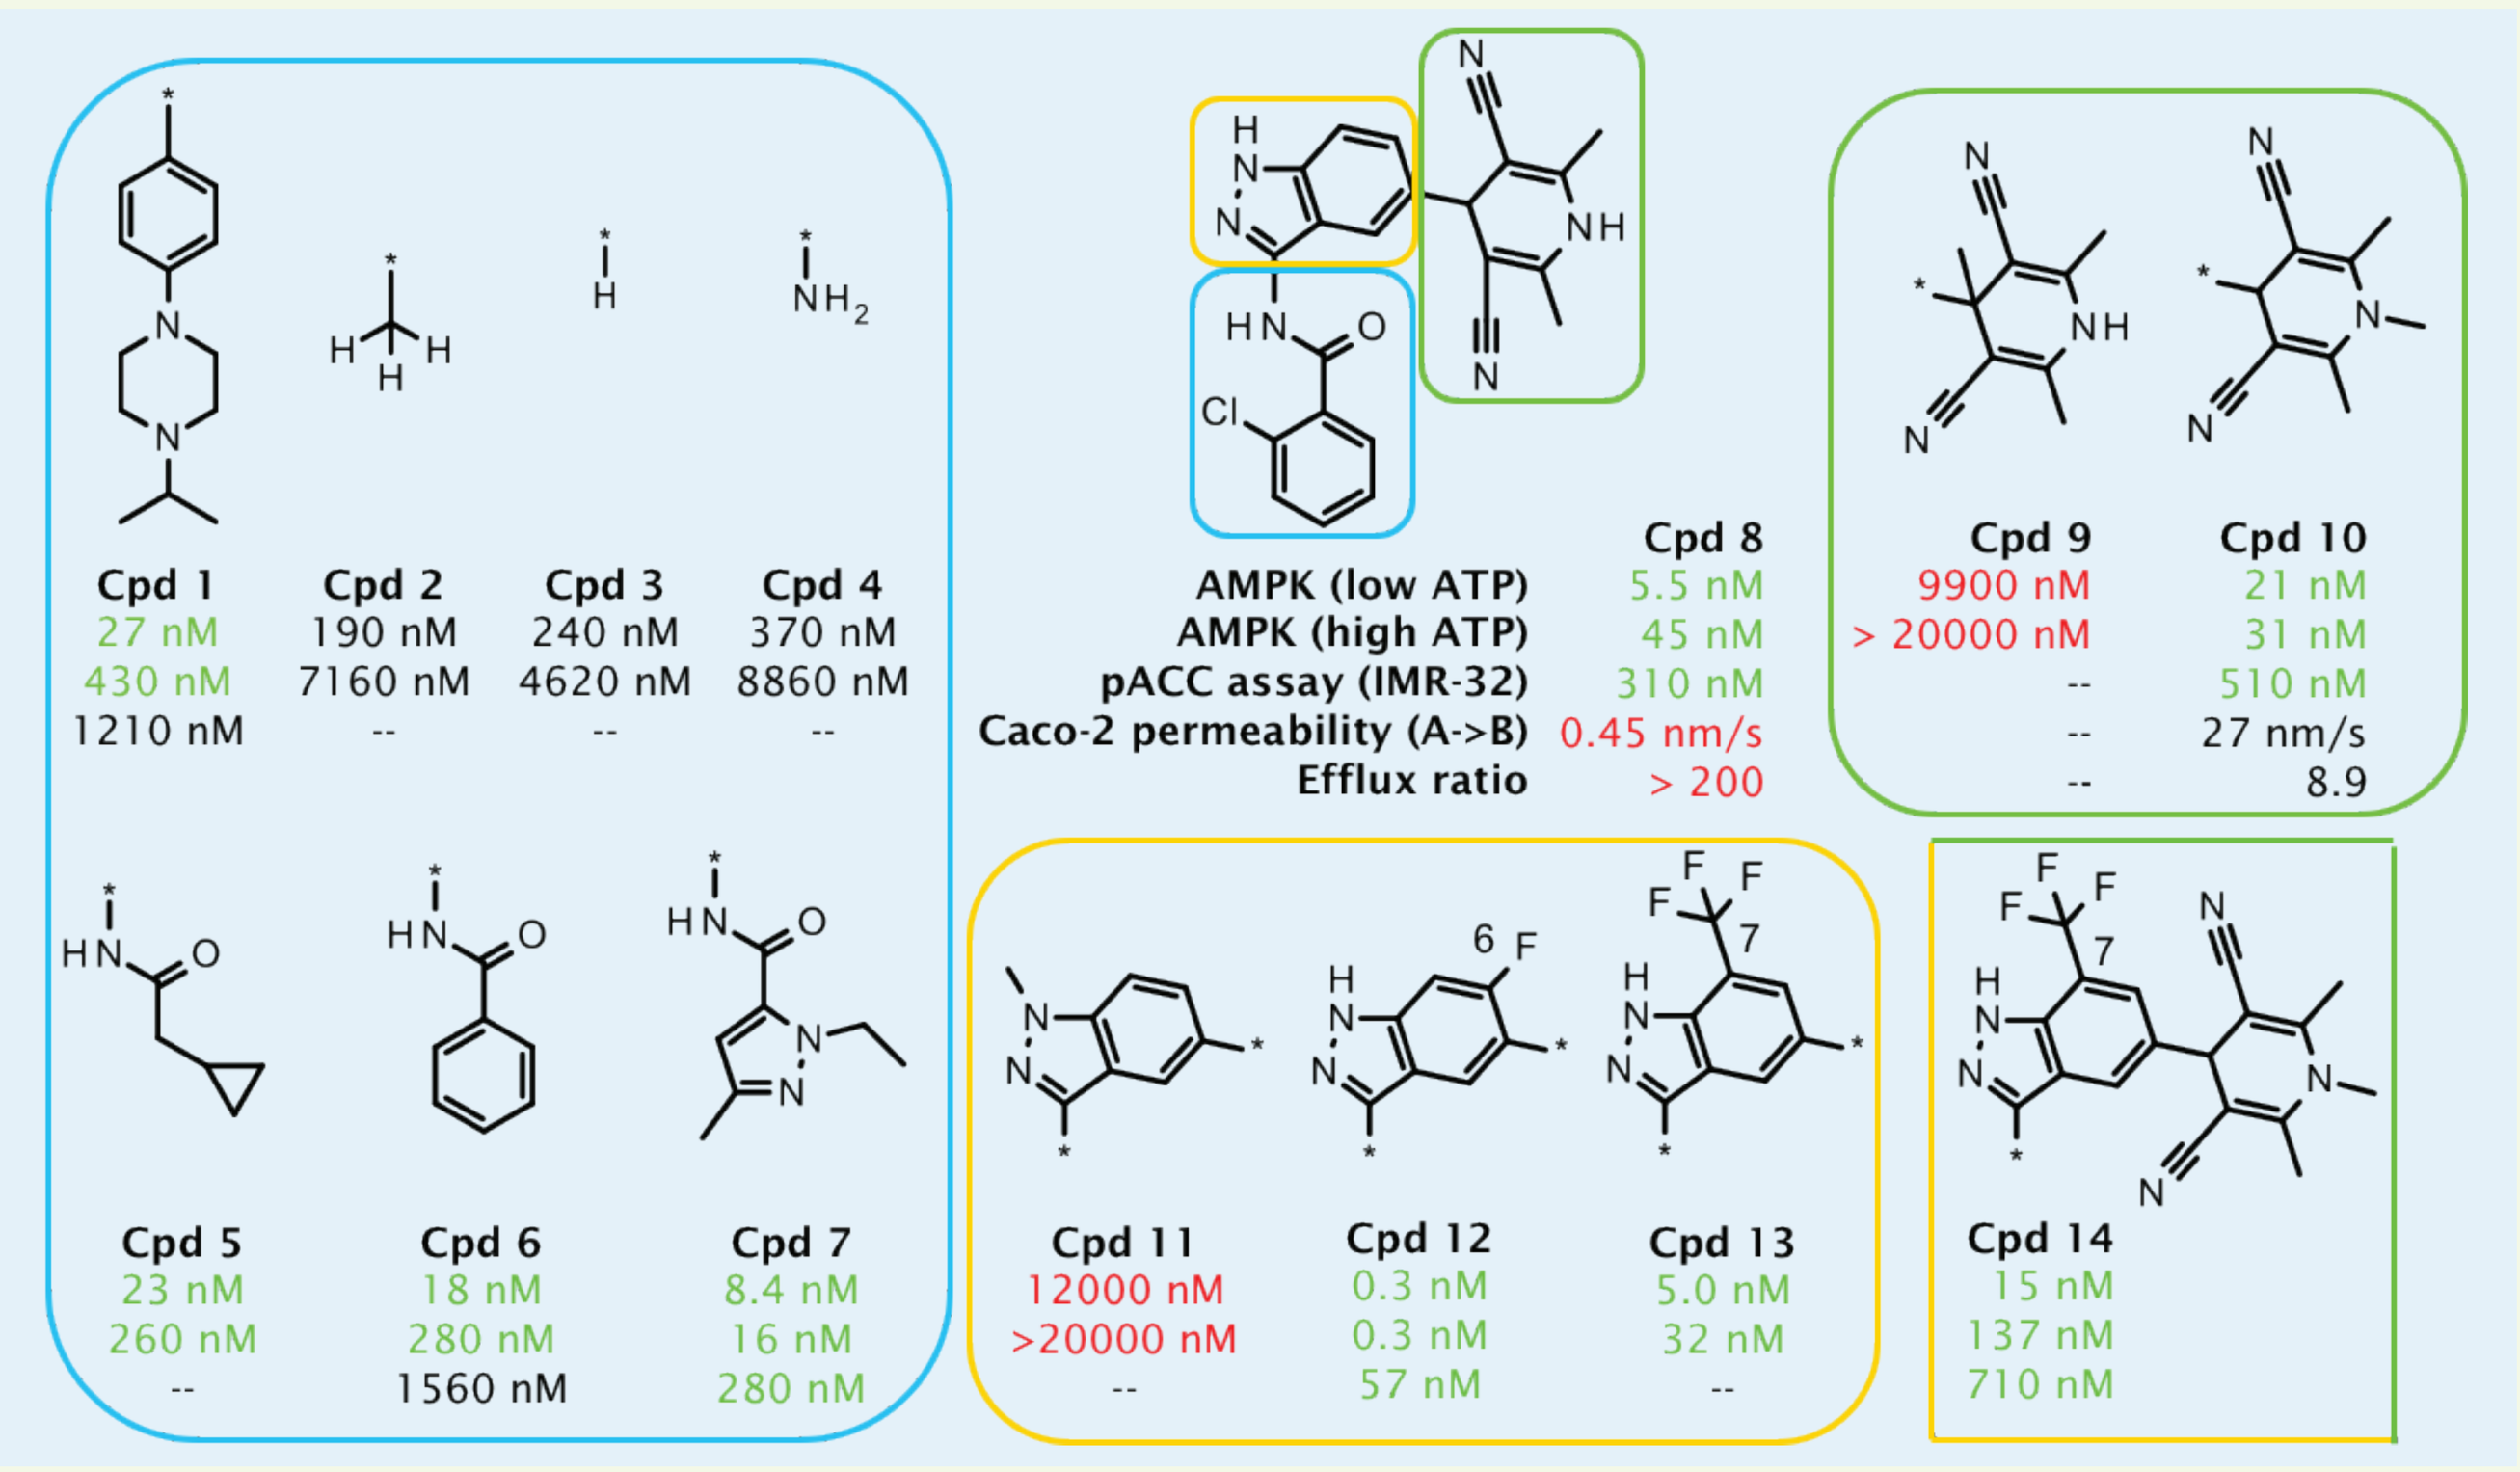
Cpd 1 27 nM 430 nM 1210 nM
Cpd 2 190 nM 7160 nM --
Cpd 3 240 nM 4620 nM --
Cpd 4 370 nM 8860 nM --
AMPK (low ATP)
AMPK (high ATP)
pACC assay (IMR-32)
Caco-2 permeability (A->B)
Efflux ratio
Cpd 8 5.5 nM 45 nM 310 nM 0.45 nm/s > 200
Cpd 9 9900 nM > 20000 nM -- --
Cpd 10 21 nM 31 nM 510 nM 27 nm/s 8.9
Cpd 5 23 nM 260 nM --
Cpd 6 18 nM 280 nM 1560 nM
Cpd 7 8.4 nM 16 nM 280 nM
Cpd 11 12000 nM >20000 nM --
Cpd 12 0.3 nM 0.3 nM 57 nM
Cpd 13 5.0 nM 32 nM --
Cpd 14 15 nM 137 nM 710 nM

Figure 2. Structure-activity relationship (SAR) of high-throughput screening (HTS) hits and first analogues as determined by AMPK kinase, cellular mechanistic pACC, and Caco-2 permeability assays. Cpd, compound; pACC, phosphorylated acetyl-CoA carboxylase.

- Amides showed improved potency (blue box), in particular 2-chloro-benzamides (compounds 8 and 12) had very high potency (**Fig. 2**).
- The F-substituent at carbon 6 (C6) of the dihydropyridine (DHP) core improved potency (compound 12, yellow box).
- Compounds with N-Me substituted DHP generally had better Caco-2 permeability and reduced efflux ratios (e.g. compound 10 vs compound 8).

X-ray structure of compound 6

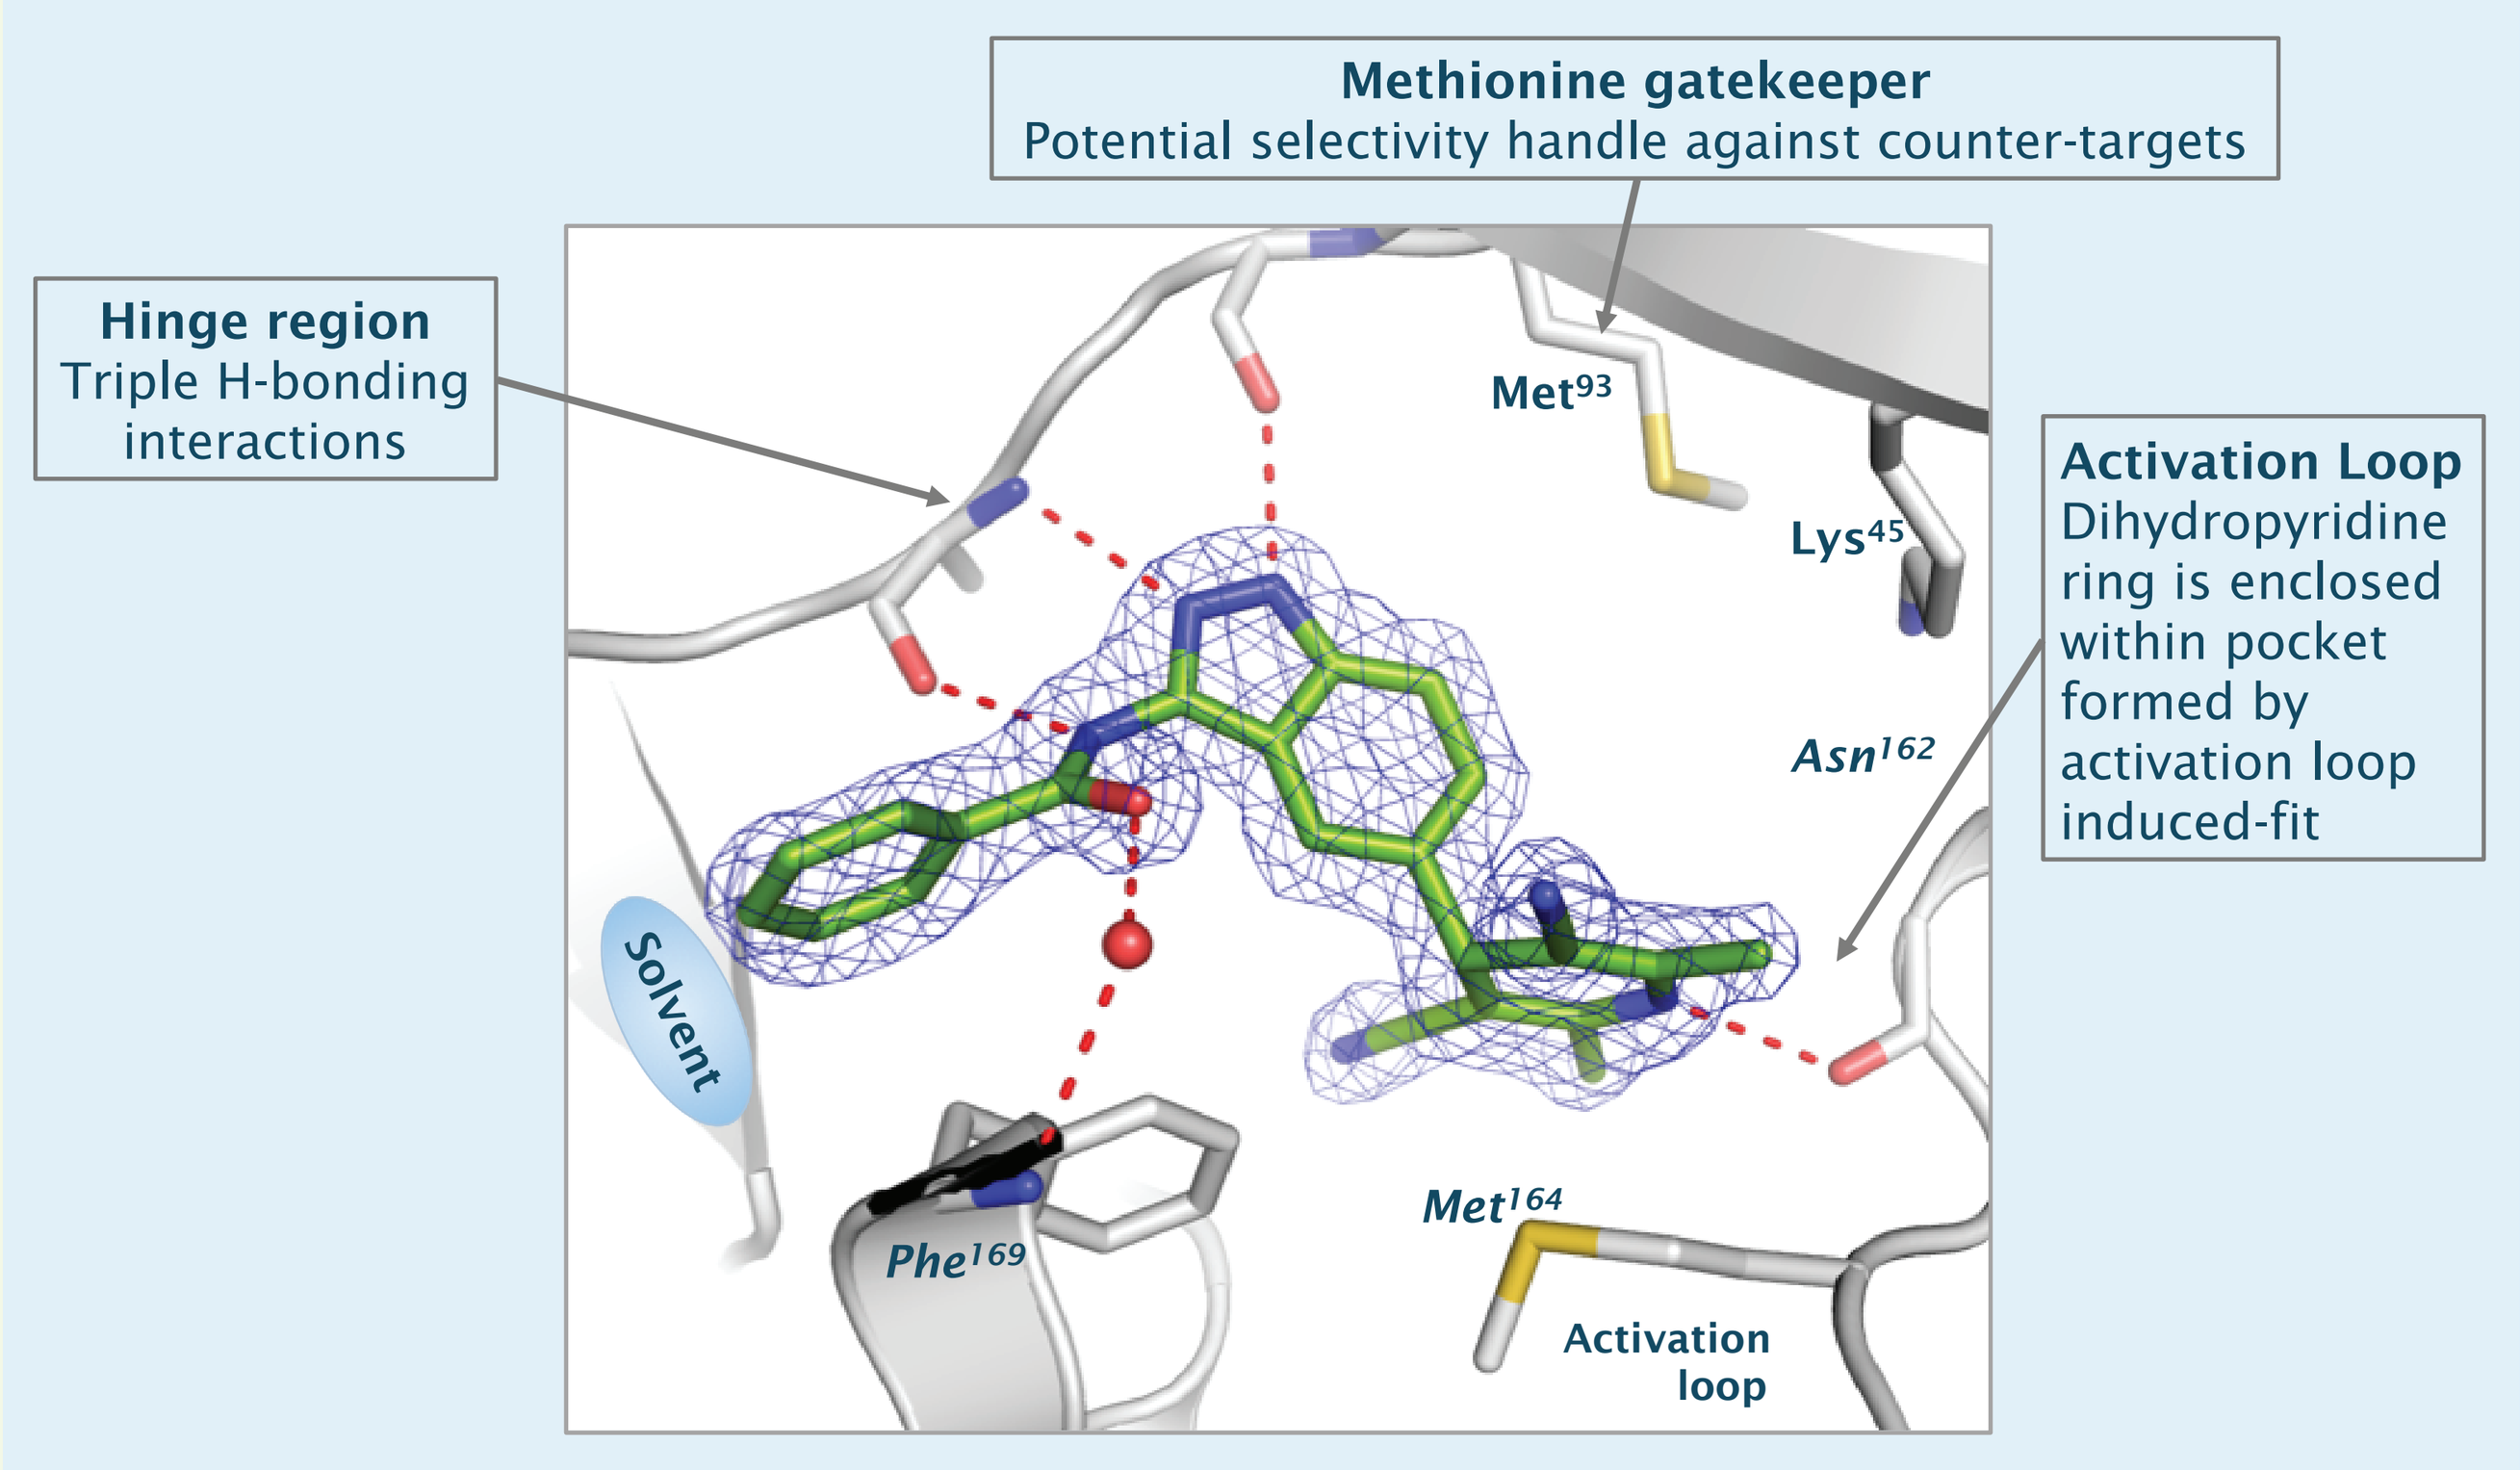
Methionine gatekeeper
Potential selectivity handle against counter-targets
Hinge region
Triple H-bonding interactions
Activation Loop
Dihydropyridine ring is enclosed within pocket formed by activation loop induced-fit
Met93
Lys45
Asn162
Solvent
Phe169
Met164
Activation loop

Figure 3. X-ray structure of compound 6. Only selected residues that form critical contacts with the compound are shown. Hydrogen bonds are depicted as dashed red lines. The complex structure was determined at 1.8 Å resolution.

- The X-ray structure of compound 6 in complex with the AMPKα2 domain allowed a better understanding of the binding mode (**Fig. 3**).

Rationale for potency improvement upon F-substitution

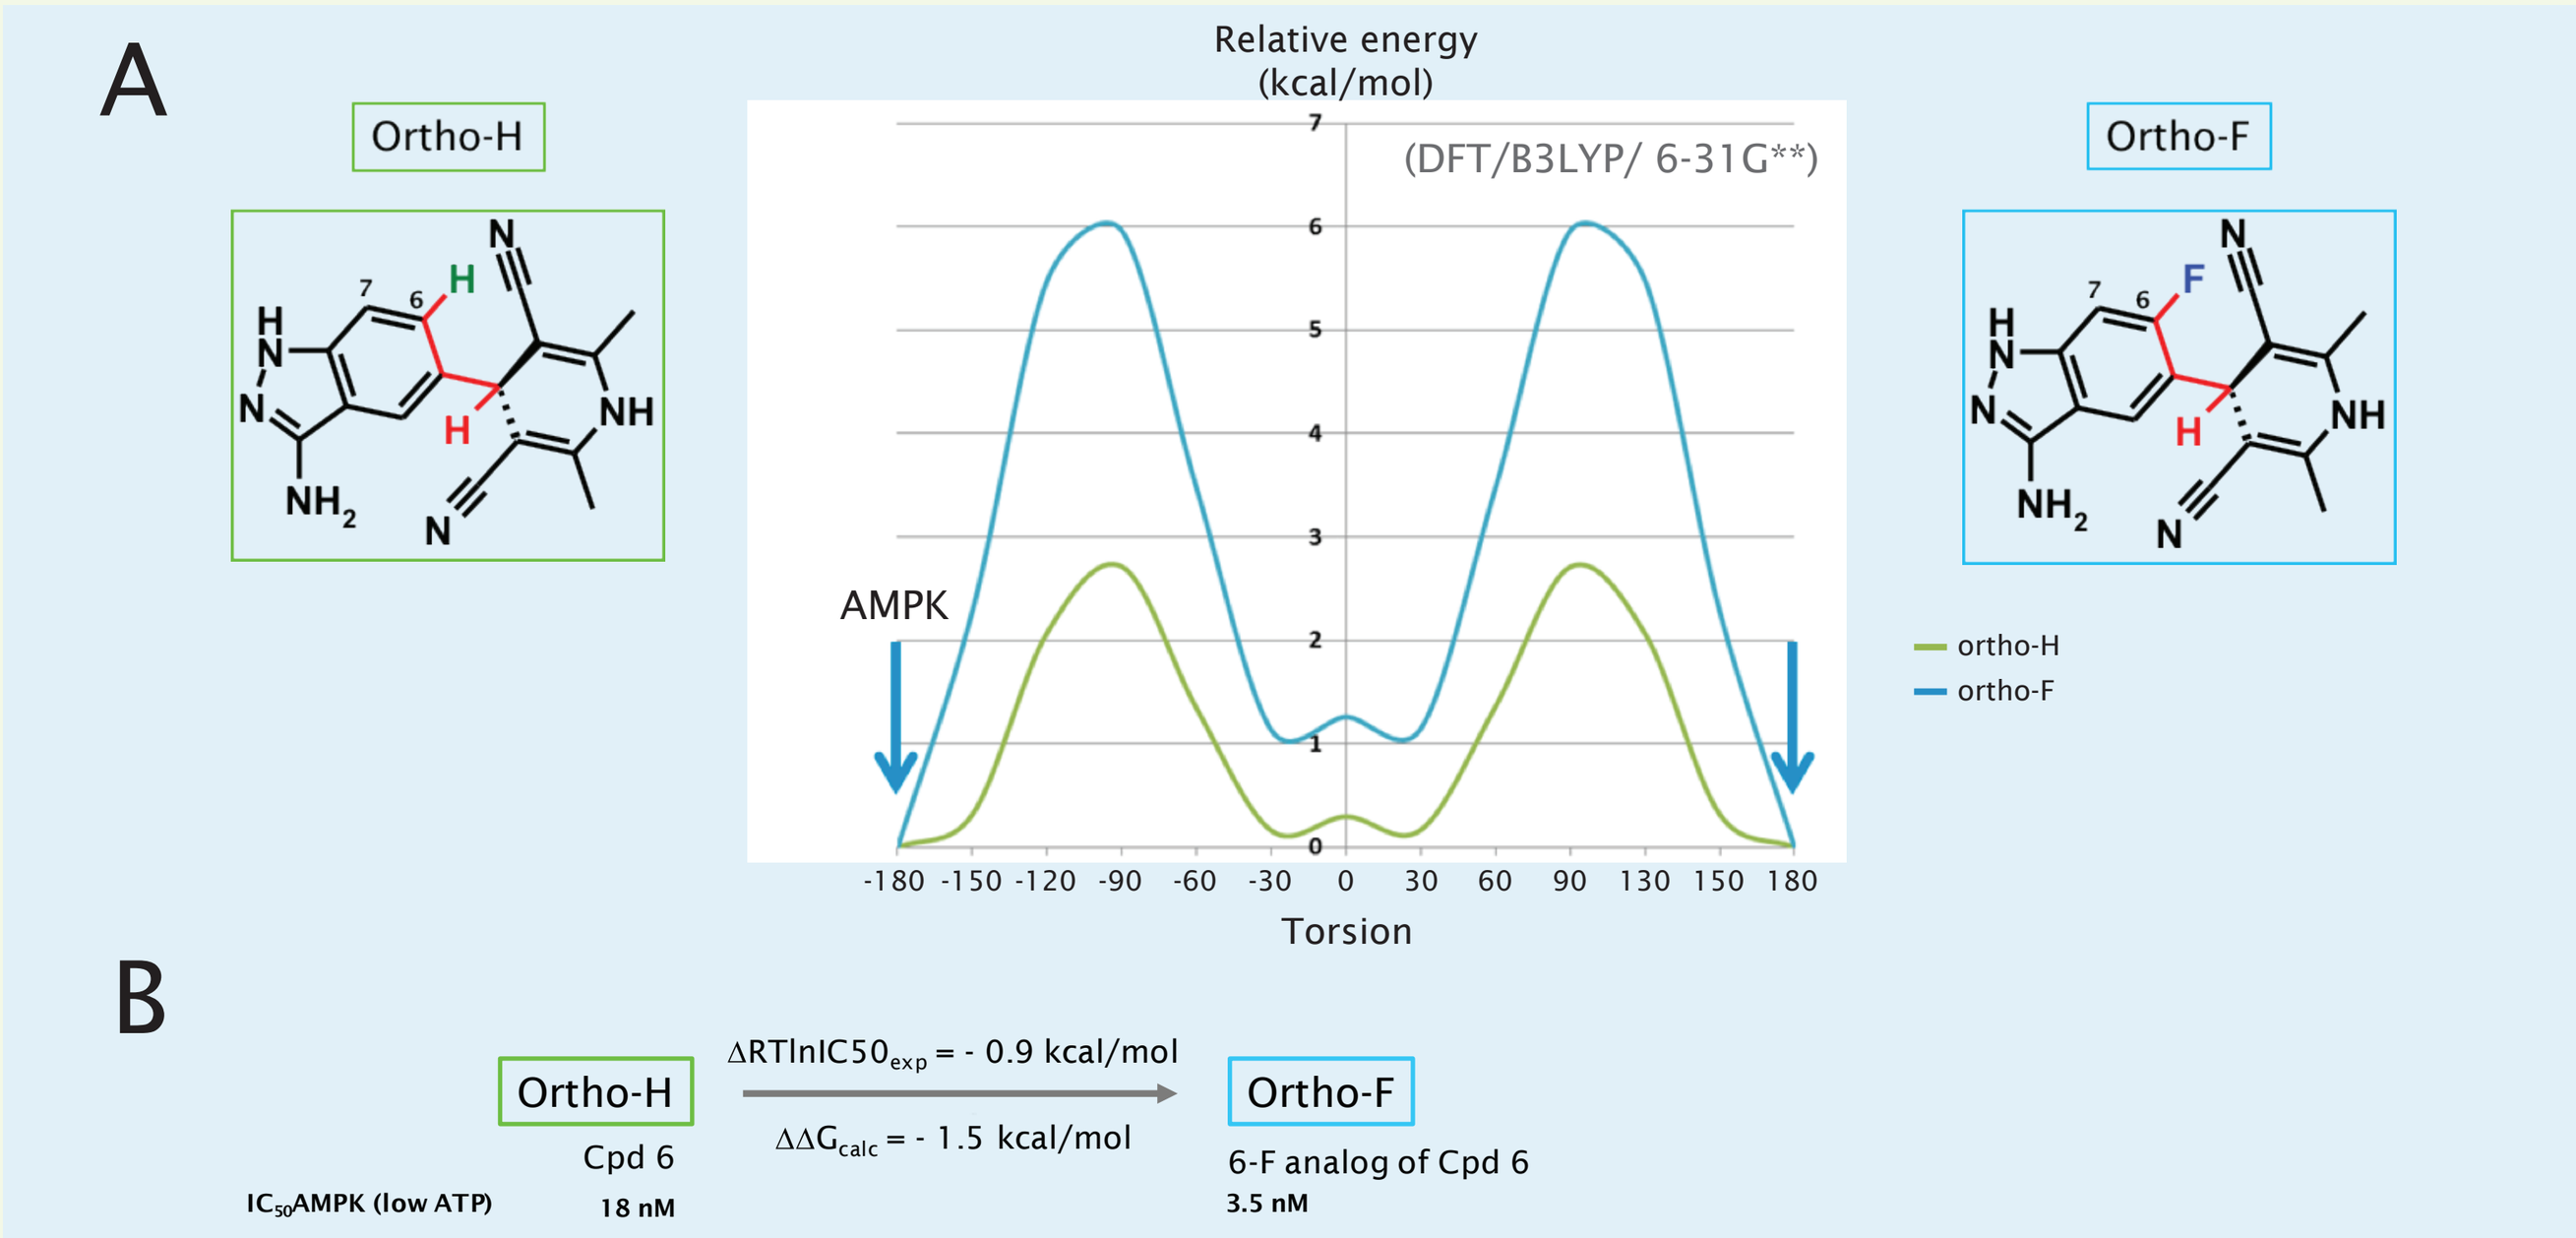
A
Ortho-H
Relative energy (kcal/mol)
(DFT/B3LYP/ 6-31G**)
AMPK
Torsion
Ortho-F
ortho-H
ortho-F
B
Ortho-H
Cpd 6
IC50AMPK (low ATP) 18 nM
ΔRTlnIC50exp = - 0.9 kcal/mol
ΔΔGcalc = - 1.5 kcal/mol
Ortho-F
6-F analog of Cpd 6
3.5 nM

Figure 4. Rationale for potency improvement upon F-substitution. (A) Conformational analysis using ab initio quantum mechanics calculations, level of theory: (DFT/B3LYP/ 6-31G**). **(B)** FEP (free energy perturbation) calculations (software: FEP/REST, Schrödinger Inc.) utilizing X-ray of **Fig. 3** to calculate free energy gain ΔΔG for 6-F analog of compound 6 compared to compound 6.

- Conformational analysis (**Fig. 4A**)
 - Trans-conformation of the core and dihydropyridine (DHP) (H-H torsion = 180°) was observed in AMPK X-ray.
 - The 6-F core (blue line) featured a much sharper and steeper energy minimum of the trans-conformation (H-F torsion = 180°) than the unsubstituted 6-H core (green line).
 - Ortho-F compounds showed higher potency compared with ortho-H compounds. This may be explained by entropical reasons, as the higher energy barrier in the ortho-F compounds locks the compounds in the right AMPK-bound conformation.
- Calculation of binding free energy (free energy perturbation, FEP) (**Fig. 4B**)
 - Potency improvement upon F-substitution could be calculated with good accuracy.

Potency and selectivity optimization of the DHP lead structure

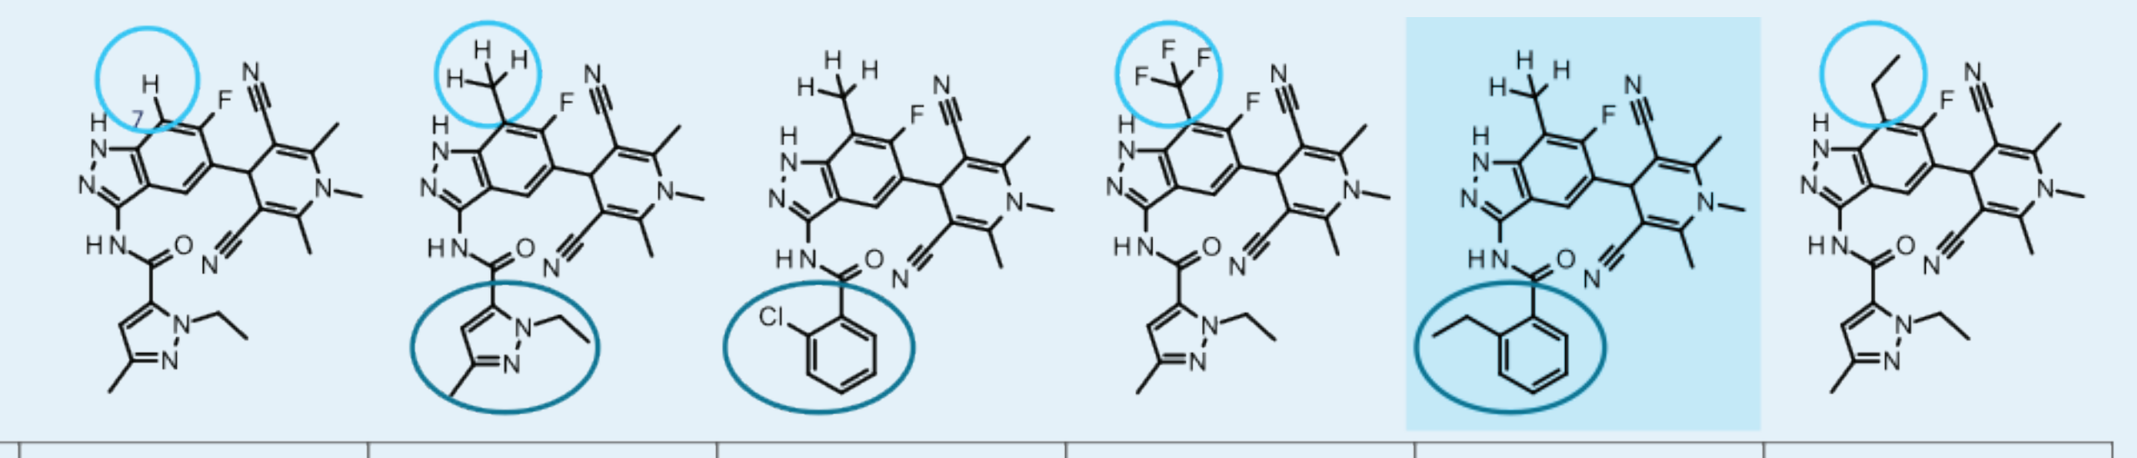

Compound No.	Cpd 15	Cpd 16	Cpd 17	Cpd 18	Cpd 19 BAY-3827	Cpd 20
AMPK, low/ high ATP [nM]	1.7 / 8.0	1.1 / 3.7	1.2 / 3.7	4.7 / 25	1.4 / 15	10 / 70
pACC (IMR-32) [nM]	43	46	97	250	150	355
Aurora A (low ATP) [nM]	14	50	89	700	480	> 10000
Ratio Aurora / AMPK	≈ 5	45	75	150	300	> 1000
Proliferation assay, IMR-32	140 nM	4200 nM	6100 nM	7600 nM	24000 nM	8860 nM

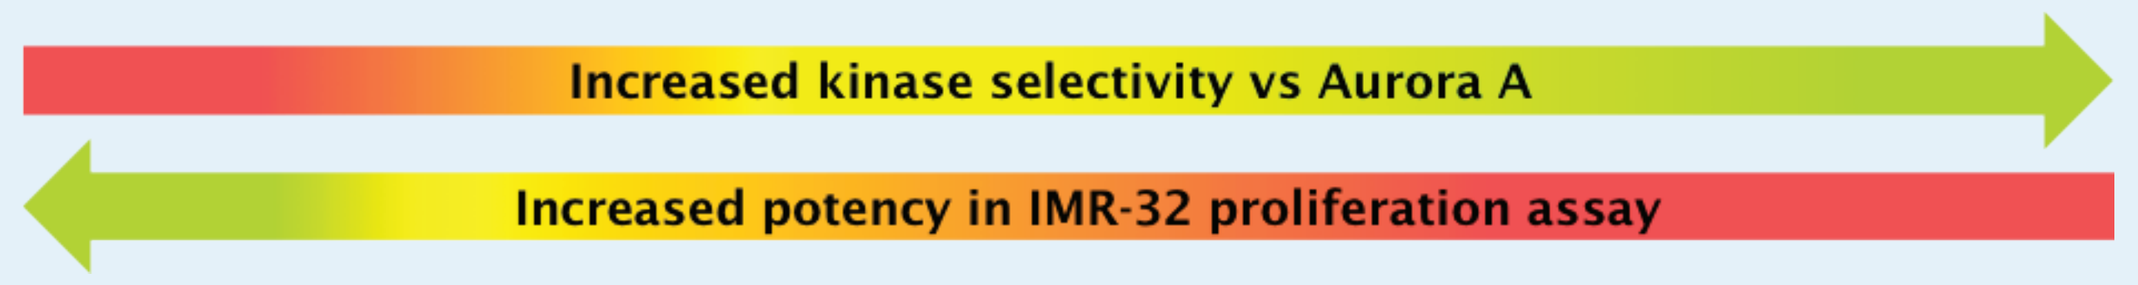
Increased kinase selectivity vs Aurora A
Increased potency in IMR-32 proliferation assay

Figure 5. Chemical optimization of the DHP lead structure with respect to potency and selectivity. Structure-activity relationship analysis for modifications at C7 of the DHP core and for modifications of the amide moiety. IC_{50} values were determined with AMPK kinase assays, with a low ATP Aurora A kinase assay, with a cellular mechanistic assay measuring phosphorylated acetyl-CoA carboxylase (pACC) and with a proliferation assay using IMR-32 neuroblastoma cells. Cpd, compound.

- The substituent at C7 of the DHP core increased the selectivity of the compounds towards Aurora kinase in the following order: -H < -Me < $-CF_3$ < Et (**Fig. 5**).
- The increased kinase selectivity of the compounds was associated with decreased potency in a proliferation assay using MYC-dependent IMR-32 neuroblastoma cells.
- Based on the high potency against AMPK and good kinase selectivity, compound 19 (called BAY-3827) was selected as a tool compound for further studies.

Potent inhibition of AMPK kinase activity does not translate into anti-proliferative activity on MYC-dependent cells *in vitro*

A

Method	Test system	Cpd 19 BAY-3827
AMPK (low/high ATP)	IMR-32	1.4 / 15 nM
Aurora A (low ATP)	IMR-32	480 nM
pACC HTRF	IMR-32	150 nM
Proliferation assay	COLO 320DM	≈ 30 µM
	LS-174T	> 30 µM
	Ramos	12 µM
	SNU-16	12 µM
	SU-DHL-10	13 µM
	Oci-Ly-7	12 µM
	JJN3	≈ 30 µM
	COLO 201, control	16 µM
	IMR-32	24 µM
	IMR-5/75	21 µM
	SK-N-F1, control	22 µM

B

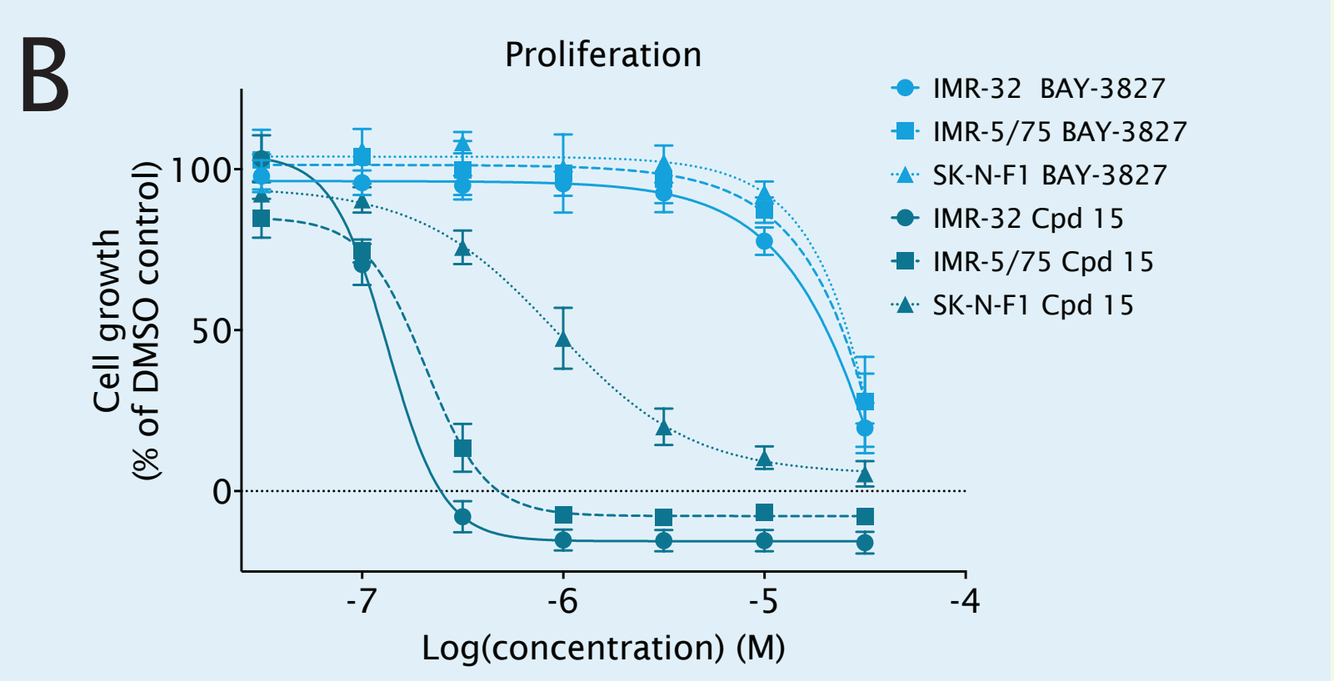
Proliferation
Cell growth (% of DMSO control)
Log(concentration) (M)
IMR-32 BAY-3827
IMR-5/75 BAY-3827
SK-N-F1 BAY-3827
IMR-32 Cpd 15
IMR-5/75 Cpd 15
SK-N-F1 Cpd 15

C

Properties of BAY-3827	
LogD @ pH 7.5	2.6
BEI / LLE (calc, AMPK low ATP)	19.5 / 6.1
S_w @ pH 6.5 [mg/L]	0.3
MW corr / TPSA [g*mol / Å²]	454 / 109
Stability (rat/human plasma, 4h) [%]	100 / 100

Figure 6. Effect of the AMPK inhibitor BAY-3827 on the proliferation of MYC-dependent cells. (A) The anti-proliferative effect of BAY-3827 was evaluated in a panel of cancer cell lines with dysregulated MYC signaling . The Colo201 and SK-N-F1 cell lines with no MYC dysregulation were used as control. IC_{50} values [µM] represent mean values determined from 2-6 individual experiments. Green/red color indicates expected/unexpected results. **(B)** The effect of BAY-3827 and compound 15 on the proliferation of different cell lines. **(C)** Selected physico-chemical and calculated properties of BAY-3827. Cpd, compound; MW, molecular weight; pACC, phosphorylated acetyl-CoA carboxylase; LogD@ pH 7.5, distribution coefficient between octanol and water at pH 7.5; BEI, binding efficiency (pIC_{50} * 1000/MW_{corr}); LLE, ligand-lipophilicity efficiency (pIC_{50} - cLogD@ pH 7.5); Sw, solubility in water; TPSA, topological polar surface area.

- The potent and selective AMPK inhibitor BAY-3827 did not inhibit cell proliferation in cancer cell lines with dysregulated MYC signaling (**Fig. 6A-B**).
- BAY-3827 was found to exhibit drug-like properties (**Fig. 6C**).

CONCLUSIONS

- A potent and selective AMPK inhibitor, BAY-3827 (**Fig. 7**), was identified through high-throughput screening and subsequent chemical optimization.
- BAY-3827-induced inhibition of AMPK did not translate into anti-proliferative activity in the various tested MYC-dependent cell lines, which is in contrast to our target validation with siRNA, and might reflect activation of alternative pathways for ATP generation.
- While the initial hypothesis of this study could not be confirmed, it can be speculated that inhibition of AMPK may have therapeutic usefulness in other biological contexts.
- The availability of potent and selective inhibitors, such as BAY-3827, may nonetheless contribute to the better understanding of the biological role of AMPK signaling in cancer.

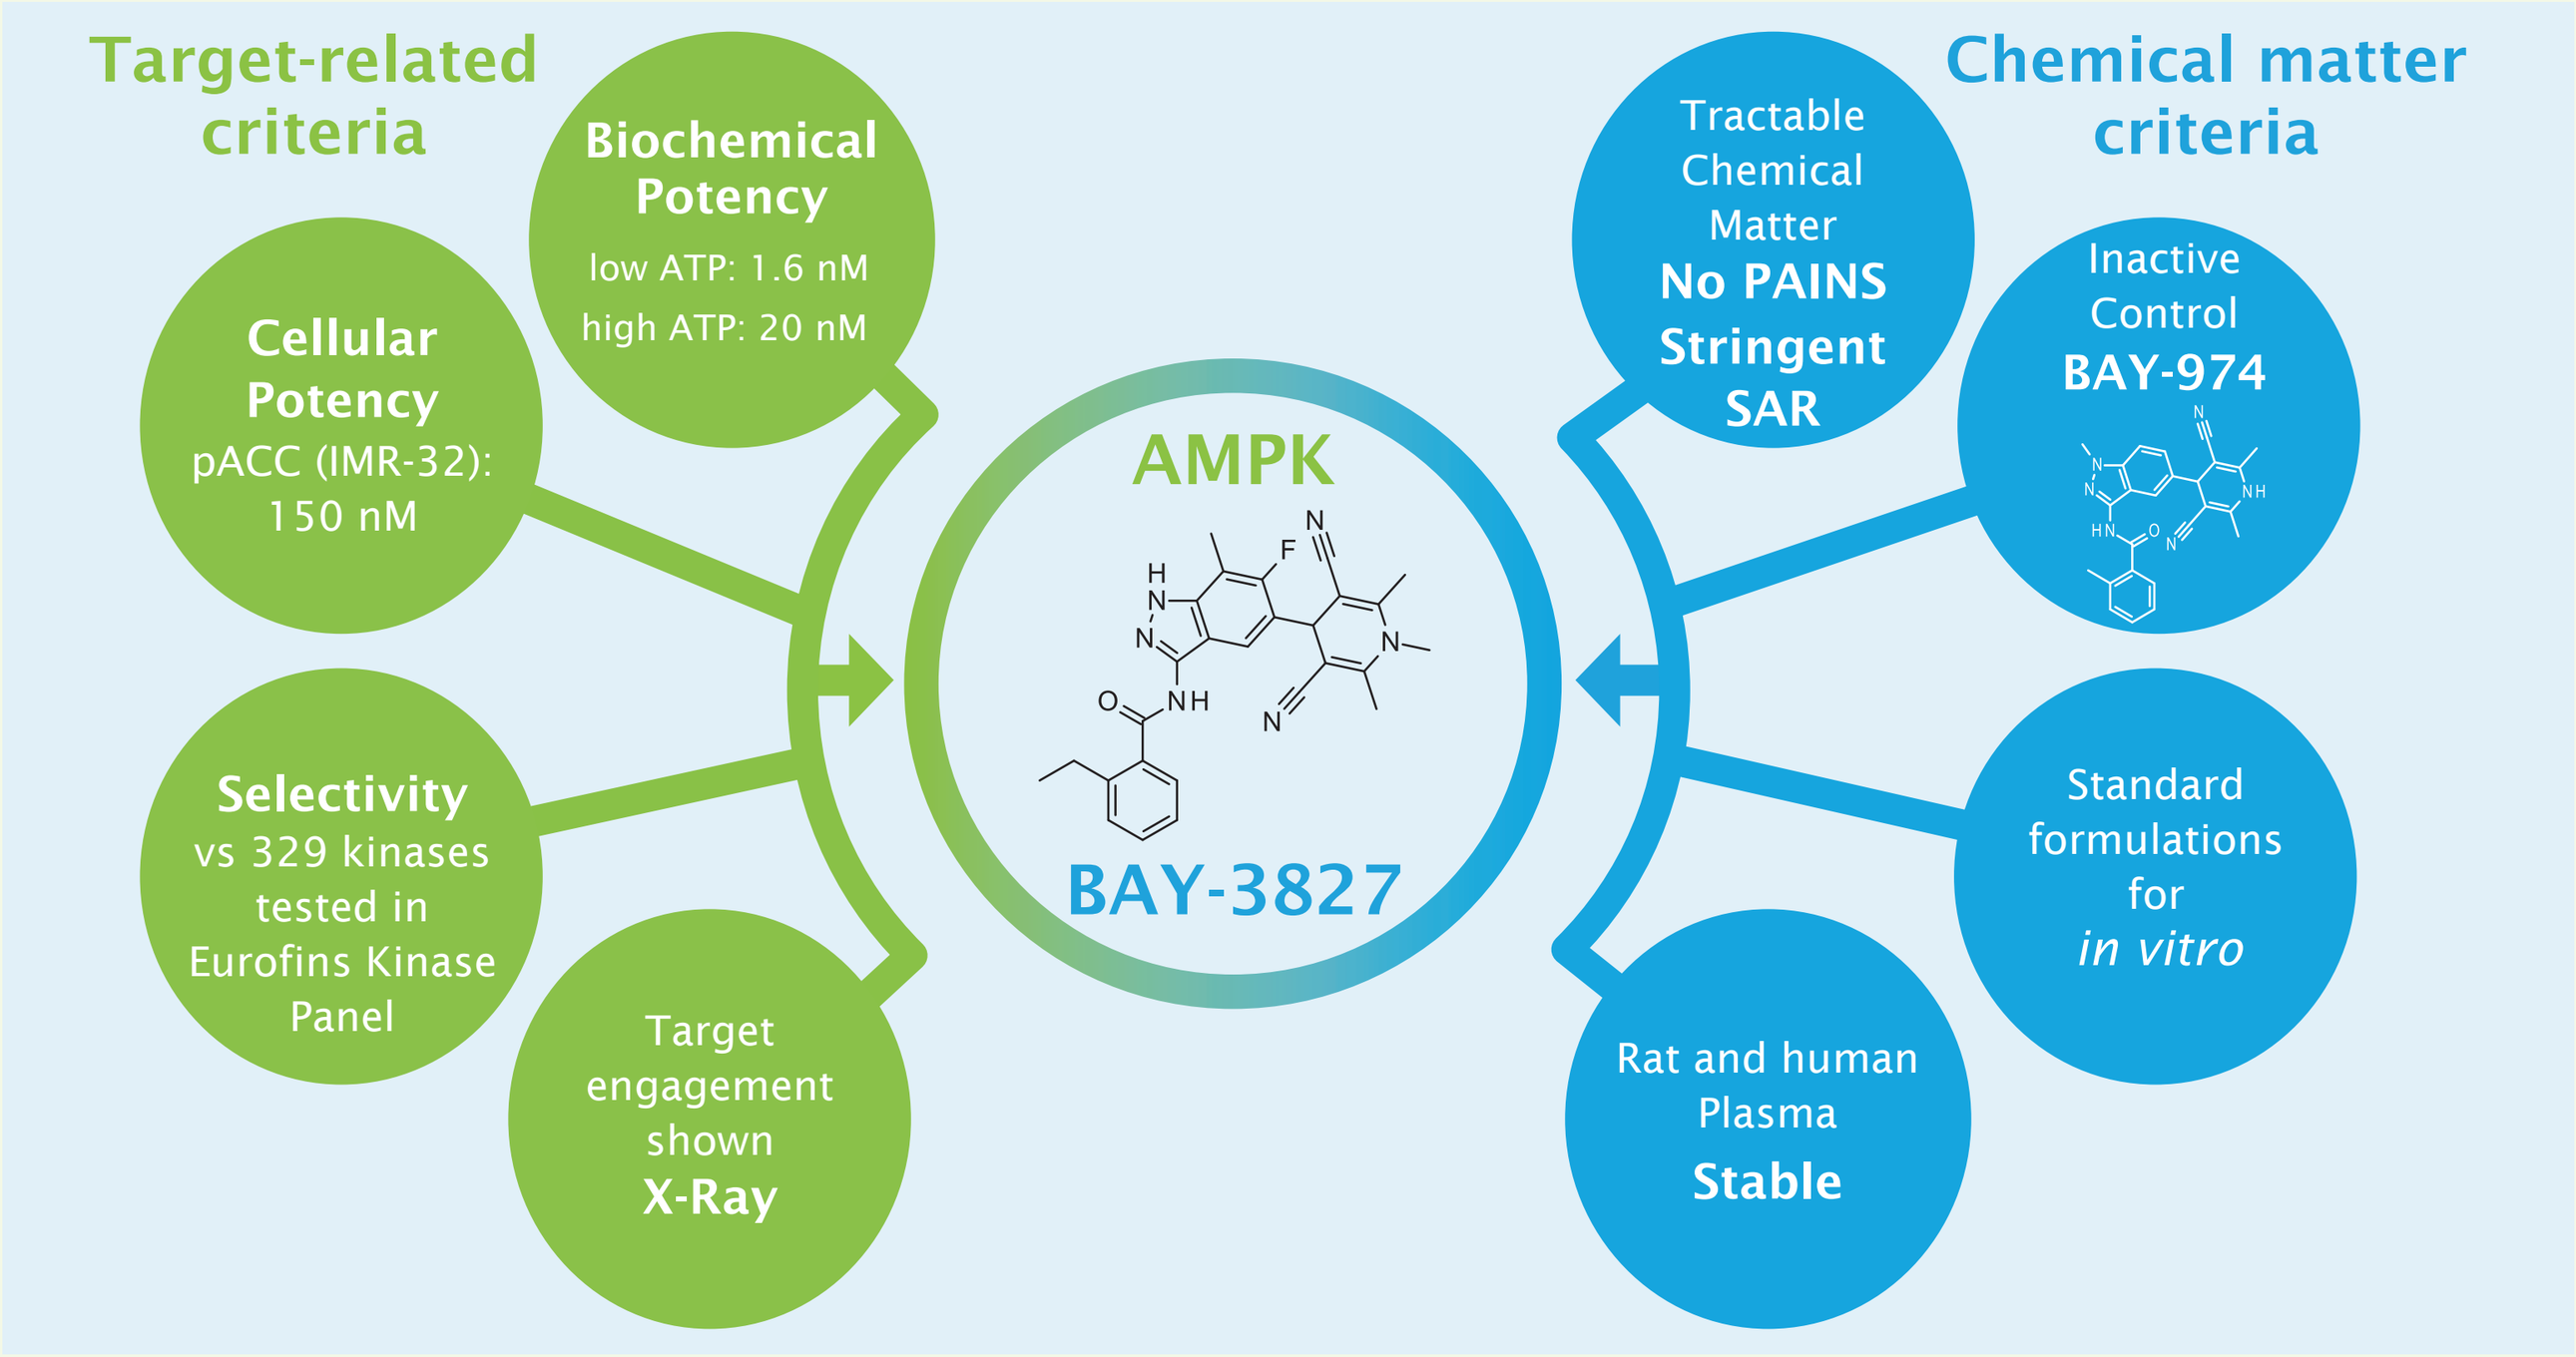
Target-related criteria
Biochemical Potency
low ATP: 1.6 nM
high ATP: 20 nM
Cellular Potency
pACC (IMR-32): 150 nM
Selectivity
vs 329 kinases tested in Eurofins Kinase Panel
Target engagement shown
X-Ray
AMPK
BAY-3827
Chemical matter criteria
Tractable Chemical Matter
No PAINS
Stringent SAR
Inactive Control
BAY-974
Standard formulations for
in vitro
Rat and human Plasma
Stable

Figure 7. AMPK-Inhibitor Probe BAY-3827.

METHODS

- AMPK kinase assays were performed at both low (10 µM) and high (2 mM) ATP concentrations. Aurora A kinase assays were performed at low ATP concentrations (10 µM).
- The cellular mechanistic assay was performed in IMR-32 neuroblastoma cells using a commercially available phospho-ACC HTRF® kit (Cisbio). This assay was used to determine the level of ACC phosphorylated at Ser79 (pACC) in cell lysates.
- Caco-2 permeability was determined with an assay using Caco-2 cells purchased from DSMZ (Germany). Permeability (Papp) was measured in the apical to basolateral (A->B) and basolateral to apical (B->A) directions. The basolateral (B) to apical (A) efflux ratio was calculated using the formula Papp (B-A) / Papp (A-B).
- Proliferation assays were performed in a panel of cells with dysregulated C-MYC (COLO 320DM, LS-174T, Ramos, SNU-16, SU-DHL-10, OCI-LY7 and JJN-3) or N-MYC (IMR-32 and IMR-5/75). Colo201 and SK-N-F1 were used as control cell lines (without C-MYC/N-MYC dysregulation).

REFERENCES

1. Faubert B *et al., Cancer Lett.* 2015;**356**;165-70.
2. Jeon SM, Hay N, *Arch Pharm Res.* 2015;**38**;346-57.
3. Liu L *et al., Nature.* 2012;**483**;608-12.

ACKNOWLEDGEMENTS

We thank Prof. Martin Eilers for scientific advice. Support on chemical syntheses by Pharmaron is gratefully acknowledged. Aurexel Life Sciences Ltd. (www.aurexel.com) is thanked for editorial assistance in the preparation of this poster, funded by Bayer AG.

Poster presented at the AACR Annual Meeting, April 14-18, 2018, Chicago, IL, USA